39

WHICH BASELINE CHARACTERISTICS INFLUENCE THE RESPONSE TO MILNACIPRAN (JONCIA®) IN PATIENT WITH FIBROMYALGIA?

Authors: O. Vitton *, P. Bunouf *, F. Bonfils *, L. Girard *

BACKGROUND

Milnacipran (MLN) has demonstrated its benefit in the treatment of patients with fibromyalgia, and obtained in November 2011 a market authorization in Australia for this indication. Fibromyalgia is a persistent pain condition that aggregates numerous symptoms (pain, fatigue, sleep disturbance, quality of life impairment…). The overall treatment effect of MLN versus placebo (PCB) observed in clinical trials was considered as moderate although clinically significant. This moderate treatment effect estimated on the whole sample of enrolled patients can be translated into a real and strong improvement in some categories of patients. The aim of this analysis is to determine the baseline characteristics defining the subgroups of patients that are associated with significantly greater treatment effect. This profiling of patients can help physician in today practice to optimize administration of Milnacipran.

METHODS

Cluster analysis, using the Ward method with Euclidian distance both for efficacy criteria and for individual patients, was first performed to exhibit natural groupings into clusters of relatively homogeneous observations. Then, an exploratory analysis was carried out using KEM (Knowledge Management and Extraction) method, which is a data mining algorithm based on association rules. Association rules methodology introduced by Agrawal and Sirikant for large database exploration [Agrawal, 1993] was further developed into various algorithms of data-mining applied to different domains [Agrawal, 1994] [Sirikant, 1995]. Since the number of generated association rules can grow exponentially, particularly when associations of attributes are used, selection of the most relevant rules is based on a range of quality measures [Berzal, 2002] [Guillet, 2007] [Kavita, 2010]. In our analysis, KEM was used to assess relationships between treatment effect on several efficacy criteria and candidate influencing factors. Totally, the 12 efficacy criteria mentioned in Table 1 were investigated, whereas the candidate influencing factors consisted of 161 baseline parameters including efficacy scores, sub-scores, and items as well as demography parameters. Continuous baseline variables were categorized into 3 categories (low, medium, high) defined according to the 33.3% and 66.6% percentiles in the whole sample. Treatment effect of MLN 100 mg versus PCB was measured in terms of odds-ratio. Subgroups were selected if the odd-ratios in the subgroups were significantly different as the overall sample ($p < 0.05$). Moreover, the minimum size of the selected subgroups was set to 5% of the total sample size.

Table 1

	EP category	List of EPs	EPs definition
Endpoints	PAIN (5)	CompRes	Composite Response
		PainResp30	Pain decrease >= 30%
		PainResp50	Pain decrease >= 50%
		PGICResp	PGIC=1.2
		PainEarlyResp30	Pain decrease >= 30% at End of dose-escalation
	Function	FIQResp50	FIQ decrease >= 50%
		PCSge6	SF36-PCS increase >= 6
		FIQRefreshResp30	FIQ Refreshing Sleep decrease >= 30%
		FIQStiffnessResp50	FIQ Stiffness decrease >= 50%
	Mental	MCSge6	SF36-MCS increase >= 6
		BDIResp50	BDI decrease >= 50%
	Fatigue	MFIResp20	MFI decrease >=20%

RESULTS

A total of 3116 fibromyalgia patients, including 836 patients administered with MLN 200 mg, 917 patients administered with MLN 100 mg, 1363 patients administered with PCB, have been investigated. These were enrolled in three three-month clinical trials: one multi-dose trial MD-02 (PCB, MLN 100 mg, MLN 200 mg) and two one dose trials GE302 (PCB, MLN 200 mg) and MD-03 (PCB, MLN 100 mg). These three clinical trials are pivotal in the TGA registration dossier.

Cluster analysis allowed a characterization of the responses on the 12 efficacy criteria. The correlation between end-point confirms the major lines of the functional categorization. We can separate, as shown in Figure 1, three symptom domains: "Pain and Global", "the Mood and central status", and "Function". It is worthwhile to note that MFIResp20 for fatigue is clustered with "Function" and should be considered as part of this domain in fibromyalgia patients. Figure 1 exhibits several subgroups of patients regarding their responses to particular efficacy criteria. The most important subgroup can be characterized by the response on "pain and global" domain, but we can observe other subgroups: Patients who respond to only Pain, only PGIC, only BDI (Mood), or only on one of the functional criteria.

Figure 1

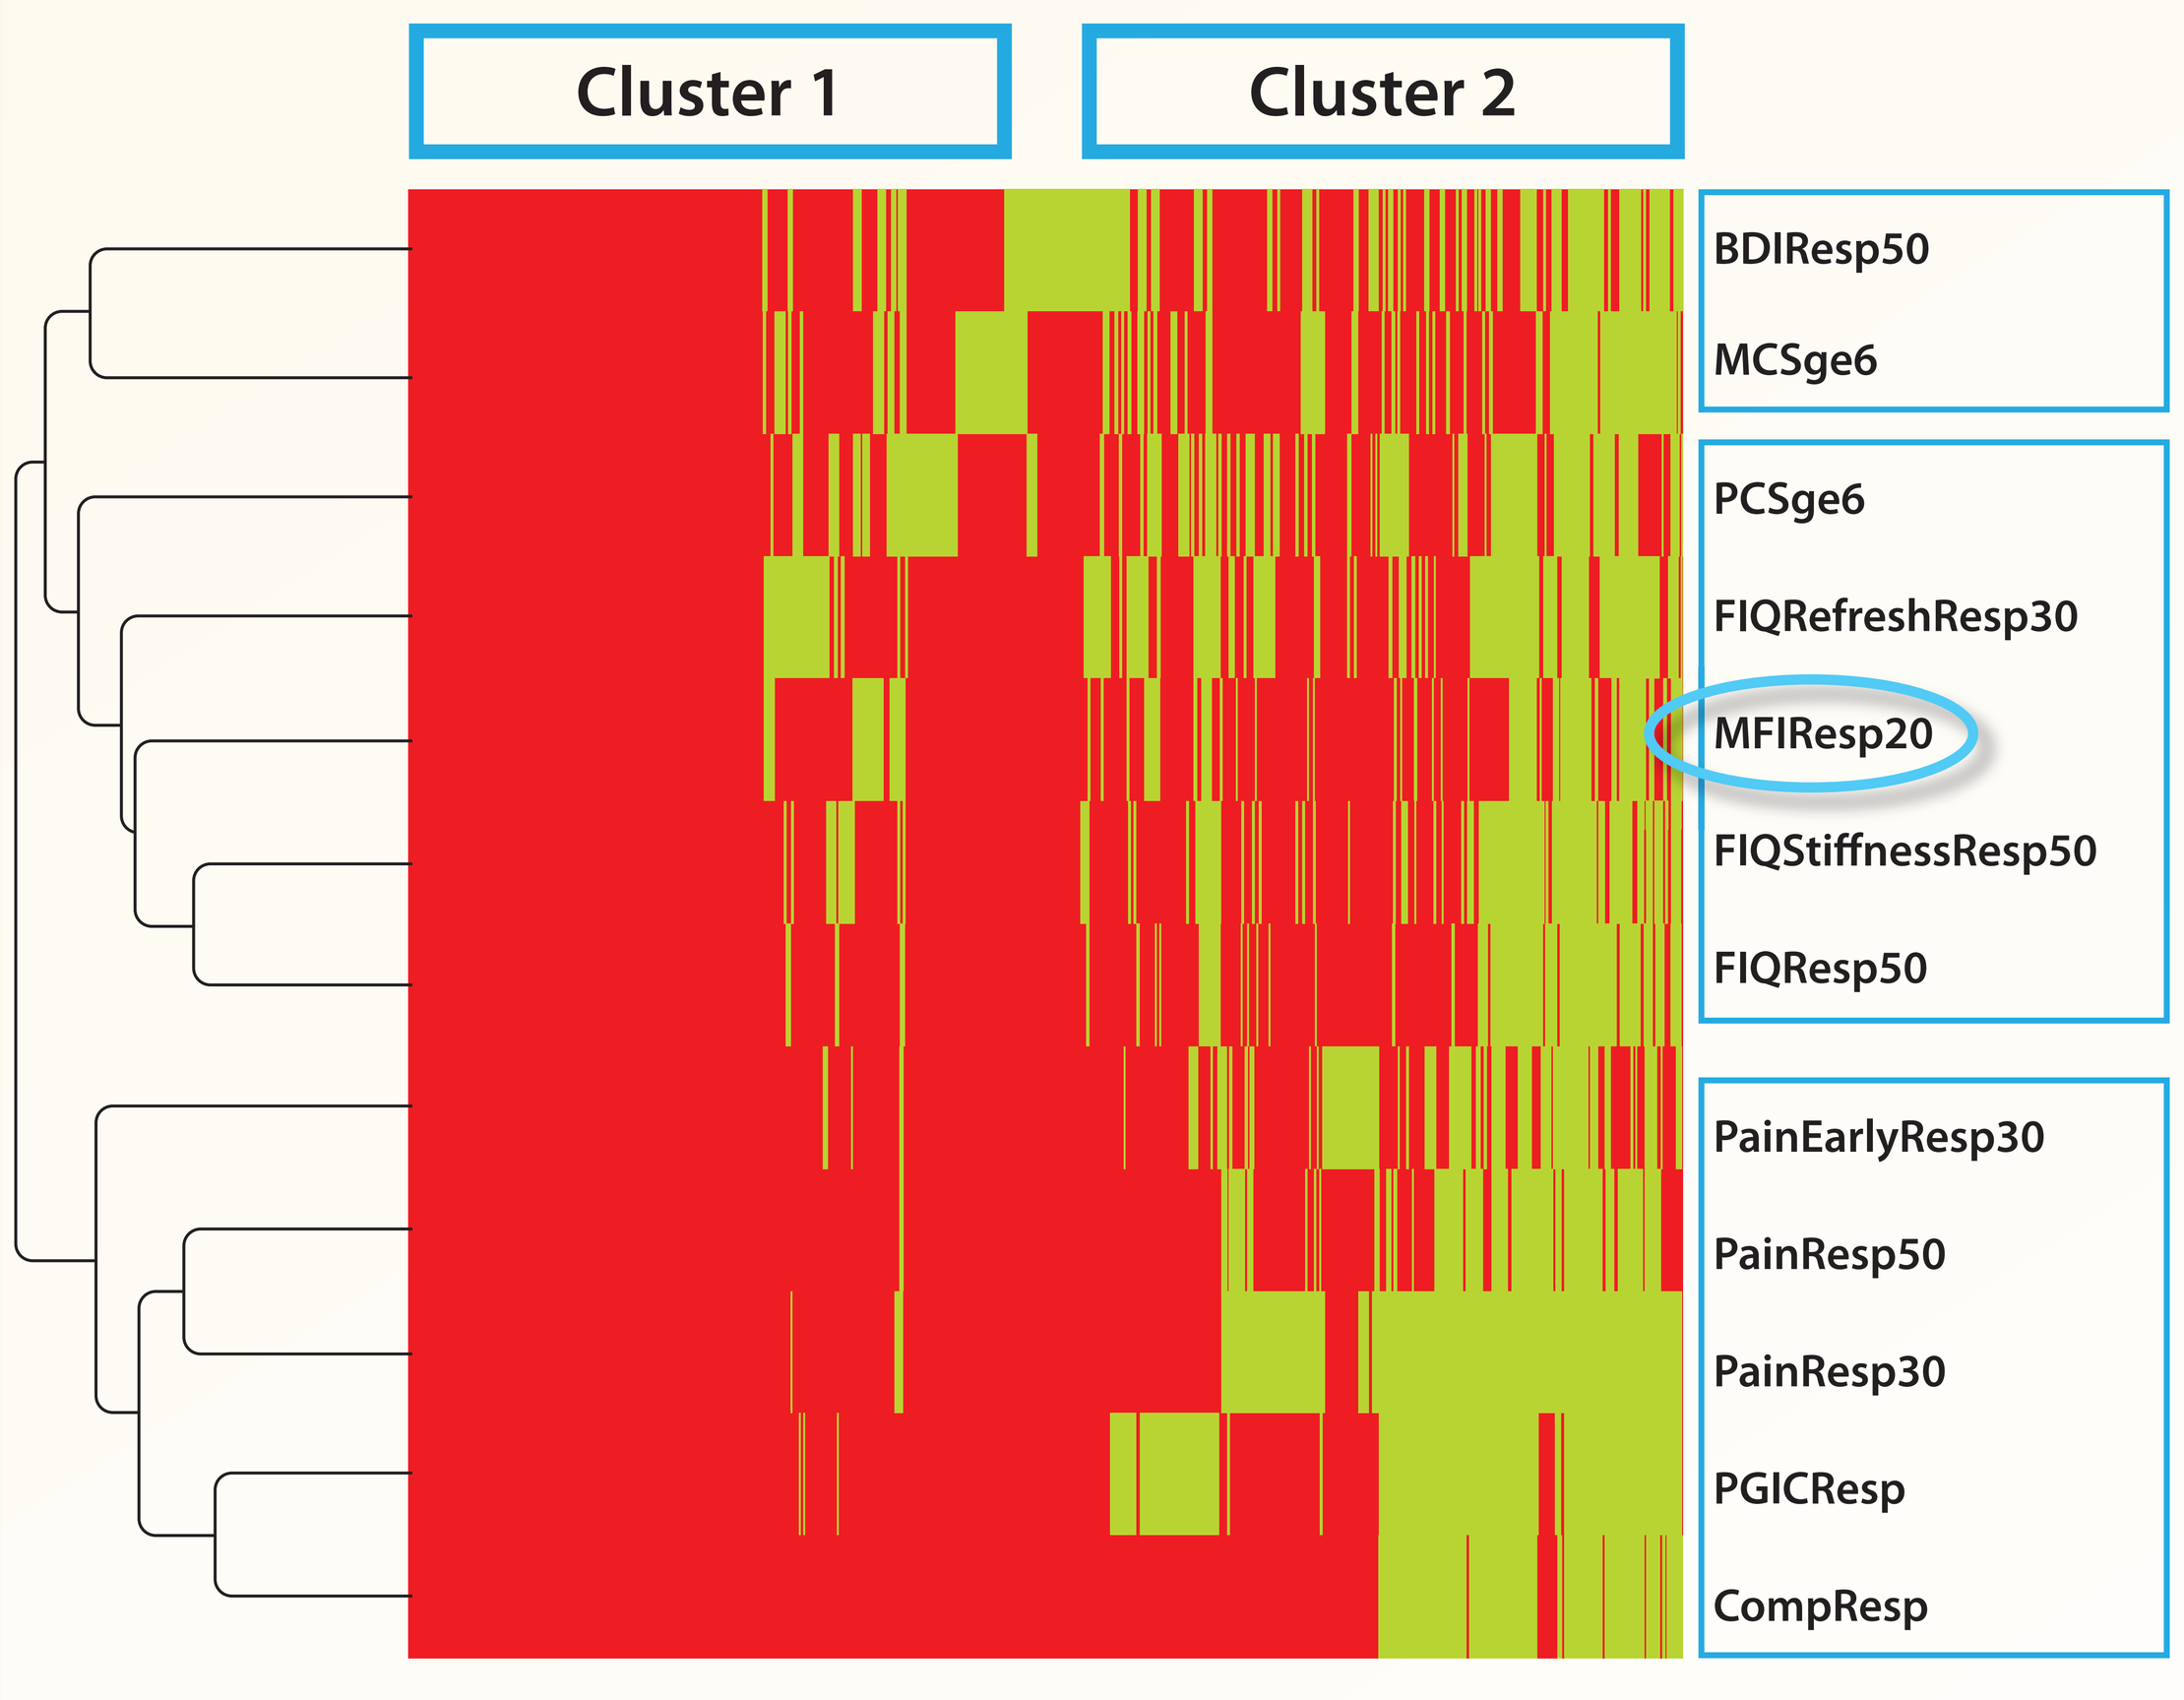
Cluster 1
Cluster 2
BDIResp50
MCSge6
PCSge6
FIQRefreshResp30
MFIResp20
FIQStiffnessResp50
FIQResp50
PainEarlyResp30
PainResp50
PainResp30
PGICResp
CompResp

Responders (R)

Non-responders (NR)

	MLN100	MLN 200	Placebo
Cluster 1 (NR)	71%	73%	82%
Cluster 2 (R)	29%	27%	18%

KEM analysis allows identification of subgroups of patients regarding their baseline characteristics. Subgroups were selected if the odd-ratios, that characterize the treatment effect of MLN versus PCB, are significantly different as the overall sample ($p < 0.05$). Moreover, the subgroup size is minimum 5% of the total sample size. A lot of parameters significantly increase the treatment effect for at least one end-point at a 5% min level of support. 8.453 baseline characters and combinations significantly the treatment effect for at least 1 of the 12 end-point, among them, 115 increase the treatment effect for at least 3 pain end-point and at least 1 of the 7 other end-point. So, different baseline characters and combinations corresponding to potential patient profiles led to a significant increase in odds-ratio measuring the treatment effect versus placebo

It is worthwhile to note that 4 subgroups based on unique baseline characters have been identified (Table 2). These are: "Pain-high", "Anxiety-low", "Change in sleeping pattern-low", and "Lifting or carrying groceries-low". In other words, when patients have high pain, are not anxious, sleep well, and have an impaired function, the odds-ratio for the composite criterion increases substantially up to 3. Moreover, Table 3 shows that the combination of characters allowed identification of several other subgroups. For example, adding to Pain other baseline parameters such as FIQ 19, BDI 16 and SF 363c can increase the odds-ratio up to 4.16 and enhance the early pain response.

Table 2

Unique of BL parameters	CompResp	PainResp50	PainResp30	PGICResp	PainEarlyResp30	FIQResp50	PCSge6	FIQRefreshResp30	FIQStiffnessResp50	MCSge6	BDIResp50	MFIResp20
Reference	1.86	1.52	1.62	2.06	1.95	1.53	1.5	1.15	1.56	1.15	1.44	1.25
Pain High: (70.64-99.5)	2.94	2.32	2.15					1.62				
FIQ19 Low: 0-21	2.77	2.16	2.13	3.09		2.36			2.32			
BDI16 Low: 0	2.88	2.17		3.03		2.21			2.43	1.73		
SF363c Low: 0	3	2.34		3.14			2.24					

Blank cell/ non-significant value (P val>0.05)

Table 3

Combinations of BL characters	CompResp	PainResp50	PainResp30	PGICResp	PainEarlyResp30	FIQResp50	PCSge6	FIQRefreshResp30	FIQStiffnessResp50	MCSge6	BDIResp50	MFIResp20	Mean OR	# MLN100 patients	% MLN patients
Reference	1.86	1.52	1.62	2.06	1.95	1.53	1.5	1.15	1.56	1.15	1.44	1.25	1.55	917	100%
Pain High: (70.64-99.5)	2.94	2.32	2.15					1.62					2.26	279	30%
BDI01 Inf: 0	3.45	2.92	2.94	3.78			2.24	1.81					2.86	185	20%
BDI02 Inf: 0	3.49	2.95	2.38	3.65				1.8					2.85	160	17%
BDI11 Inf: 0	3.51	2.93	2.6	3.41				1.68					2.83	161	18%
BDI13 Inf: 0	3.48	2.79	2.5	3.18	3.37		2.11	1.85					2.75	185	20%
MASQ02 Med: 2	5.14		3.03	4				2.21					3.60	134	15%
SF3611b Med: 25-50	4.09		2.79	3.26				2.25					3.10	154	17%
FIQ19 Low: 0-21	2.77	2.16	2.13	3.09		2.36			2.32				2.47	355	39%
BDI14 Inf: 0	3.04	2.4	2.22	3.43		2.61			2.6				2.72	320	35%
BDI16 Low: 0	5.01		2.85	4.98					3.8				4.16	127	14%
MASQ02 Med: 2	4.52	3.51	2.94	3.73				2.04	3.27				3.34	140	15%
MFI RM Low: (4-9)	3.01	2.76	2.64	3.51					2.56				2.90	159	17%
Pain Low: (21.21-57.79)	3.04		2.44	3.43				1.79	3.33				2.81	144	16%
SF36 MCS High: (51-75.47)	3.23	2.51	2.74	3.37		2.49			2.43				2.80	214	23%
SF36 PCS Low: (11.785-30.865)	3.58		2.83	3.7	3.39			1.91					3.08	123	13%
Weight High: (85.3-198.9)	3.61	2.74		3.34					2.87				3.14	137	15%
BDI16 Low: 0	2.88	2.17		3.03		2.21			2.43	1.73			2.41	127	14%
BDI02 Inf: 0	3.35	2.47	2.34	3.54		2.49			2.88			1.9	2.71	110	12%
BDI11 Inf: 0	3.15	2.33		3.25					4.21				3.24	115	13%
MASQ20 Med: 2-3	3.31	2.45		3.46		3.08			3.08				3.08	83	9%
MASQ22 Med: 2-3	2.93	2.45		3		2.78	2.19		2.92				2.71	96	10%
SF3611c Med: 50-75	3.23		2.5	3.27		2.78			2.44			2.05	2.71	84	9%
SF364b Med: 25-50	3.78	2.64		3.93					2.76				3.28	92	10%
SF364c Med: 25-50	3.42	2.76		3.36		2.84			2.35			2.21	2.82	82	9%
SF369h Med: 50-75	3.43	2.72		3.91		3.07	2.15		3.7	1.98		2	2.87	91	10%
SF363c Low: 0	3	2.34		3.14			2.24						2.68	195	21%
BDI06 Inf: 0	3.08	2.4		3.05			2.41						2.74	166	18%
SF362 Med: 25-50	3.26	2.7		3.13			2.26						2.84	154	17%

CONCLUSION

KEM analysis exhibited associations among efficacy criteria and existence of domains in fibromyalgia patients. KEM also identified subgroups of patients representing at least 5% of the whole sample size and associated with substantially greater treatment effect of MLN 100 mg versus PCB ($p < 0.05$). So, it has been possible to determine patient profile defining subgroups associated with odds-ratios over 4.

Further investigations should be conducted especially to explore the dose 200 mg. However, this analysis already confirms the benefit of Milnacipran in several representative categories of patients and justifies its use for treating FM patients.

References:

**Pierre Fabre, Research & Development center, Toulouse, France*

- *[Agrawal, 1993] Rakesh Agrawal, Tomasz Imielinski and Arun Swami (1993) Mining association rules between sets of items in large databases. ACM SIGMOD Record, 22,2: 207-216.*
- *[Agrawal, 1994] Rakech Agrawal and Ramakrishnan Srikant (1994) Fast Algorithms for mining association rules. Proccedings of the 20th VLDB conference.*
- *[Srikant, 1995] Srikant R., and Agrawal R. (1995) Mining Generalized Association Rules. procceding of the 21st Int'l Conference on Very Large Databases*
- *[Berzal, 2002] Fernando Berzal, Ingnacio Blanco, Daniel Sanchez and Maria-Amparo Vila (2002) Measuring the accuracy and interest of association rules: a new framework. Intelligent Data Analysis,6, 221-235.*
- *[Guillet, 2007] Fabrice Guillet and Howard J. Hamilton (2007) Quality measures in data mining. Ed. Springer.*
- *[Kavita, 2010] Kavita Das and O.P. Vyas (2010) A suitability study of discretization methods for associative classifiers. International Journal of Computer Applications (0975-8887), 5, 10, 46-51*

Poster presented at the APS (Australian Pain Society) 2013